AACR D3 2026
Poster A082

AP3-guided biological SAR enables discovery of ACR-2316, a novel clinical-stage WEE1/PKMYT1 inhibitor designed for CDK1/2 and PLK1 pathway activation

Lei Shi[1], Marc Isaksson[2], Ahmed Youssef[1], Valentina Siino[2], Ignacio Arribas Diez[2], Bryan Thornlow[1], Nina Lipjankic[2], Ruban Cornelius[2], Zachary Best[1], Martina Pasetto[2], Jessica Hopkins[2], Mohamed Eldeeb[2], Anna-Maria Alves[1], Portia Lombardo[1], Taronish Dubash[1], Maria Rodriguez-Zabala[1], Corey Xu[1], Georgia Mitsa[2], Subodh Kumar[1], Kate Rappard[1], Joelle Baddour-Sousounis[1], Ayesha Murshid[1], Magnus E. Jakobsson[2], Reina Improgo[1], Helen Nilsson[2], Michail Shipitsin[1], Caroline Wigerup[2], Christina Scherer[1], John van Duzer[1], Joon Jung[1], David A. Proia[1], Kristina Masson[1], Peter Blume-Jensen[1]

[1]Acrivon Therapeutics Inc., Watertown, MA, USA, [2]Acrivon AB, Medicon Village, Lund, Sweden

Download this poster

INTRODUCTION

WEE1 and PKMYT1 regulate G2/M progression and DNA damage response through inhibitory phosphorylation of CDK1/2. Using Acrivon Predictive Precision Proteomics (AP3), we mapped the intracellular pathway alterations after WEE1 and PKMYT1 inhibition and translated compensatory signaling into biological SAR criteria. AP3 revealed reciprocal CDK1 T14/Y15 regulation and quenching of WEE1 inhibitor-induced phosphosites by PKMYT1 inhibition, supporting dual WEE1/PKMYT1 inhibition. This strategy guided discovery of ACR-2316, a selective WEE1/PKMYT1 inhibitor designed for potent single-agent pro-apoptotic tumor cell killing through CDK1/2 and, importantly, PLK1 pathway activation. AP3 profiling prioritized ACR-2316 over other lead candidates by demonstrating desired signaling pathway effects not discernible from binding/selectivity assays alone.

Approach

AP3 platform transcends the boundaries of traditional drug discovery with biological SAR

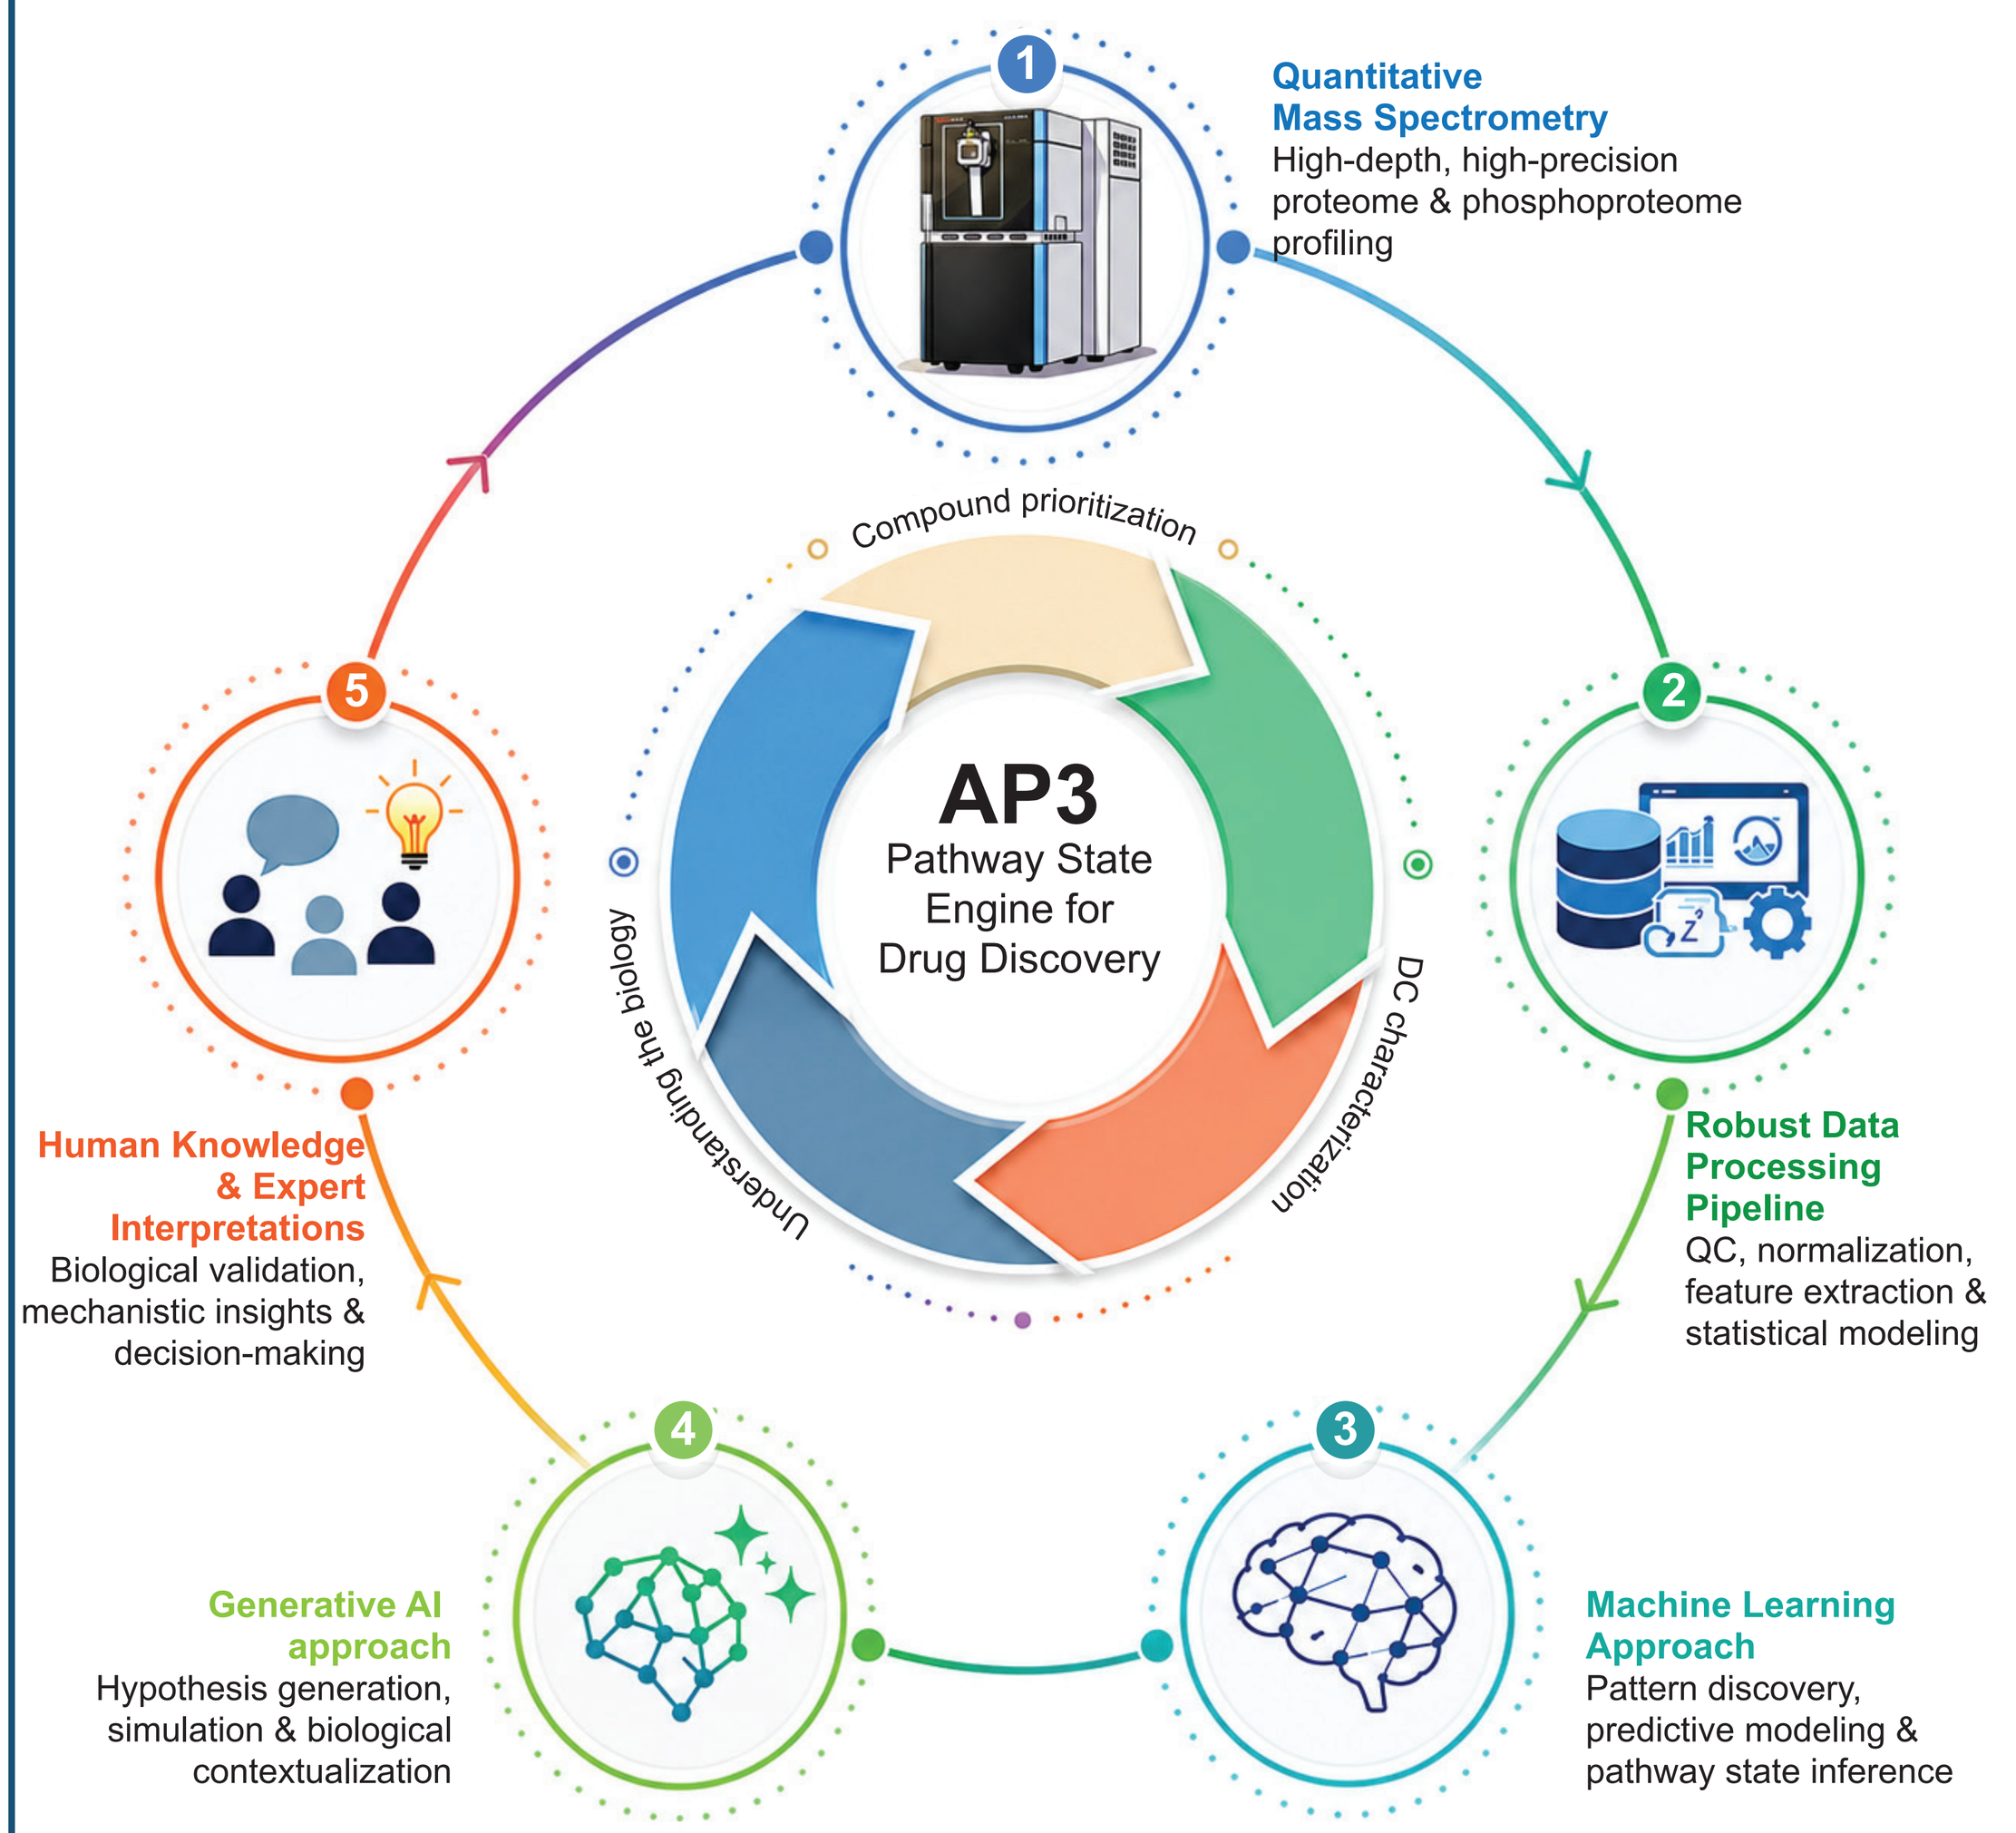
AP3
Pathway State Engine for Drug Discovery
Compound prioritization
DC characterization
Understanding the biology
1 Quantitative Mass Spectrometry
High-depth, high-precision proteome & phosphoproteome profiling
2 Robust Data Processing Pipeline
QC, normalization, feature extraction & statistical modeling
3 Machine Learning Approach
Pattern discovery, predictive modeling & pathway state inference
4 Generative AI approach
Hypothesis generation, simulation & biological contextualization
5 Human Knowledge & Expert Interpretations
Biological validation, mechanistic insights & decision-making

- Acrivon's Predictive Precision Proteomics (AP3) integrates high-throughput quantitative phosphoproteomics with artificial intelligence, machine learning, and pathway-based interpretation to enable biology-guided structure–activity relationship (SAR) analysis.
- AP3 profiled internal and competitor compounds throughout Acrivon's discovery workflow to guide compound optimization and selection.
- AP3-defined intracellular pathway signatures enabled prioritization of ACR-2316 based on biological activity beyond kinase selectivity alone.
- AP3 further characterized ACR-2316, confirming robust regulation of the desired biological pathways and supporting its advancement into clinical development.

CONCLUSIONS

- AP3 is a novel platform enabling rational design of molecules based on desirable intracellular pathway effects subsequent to traditional *in vitro* drug discovery efforts.
- AP3 established a biological SAR based on compensatory WEE1/PKMYT1 signaling and enabled mechanism-driven prioritization of ACR-2316 by capturing network-level biology beyond traditional binding and kinase selectivity assays.
- ACR-2316 is currently being evaluated in a Phase I/2 clinical trial (NCT06667141) and has demonstrated encouraging clinical antitumor activity, including durable PRs and tumor shrinkage, in lung cancer patients combined with a favorable safety profile.

RESULTS

1. AP3 Biological SAR, Part 1: Defining WEE1/PKMYT1 Mitotic Signaling and Synergy-Associated Phosphorylation to Guide Development Candidate Selection

AP3 profiling and analysis of mitotic signaling altered by Adavosertib, Lunresertib and their combo

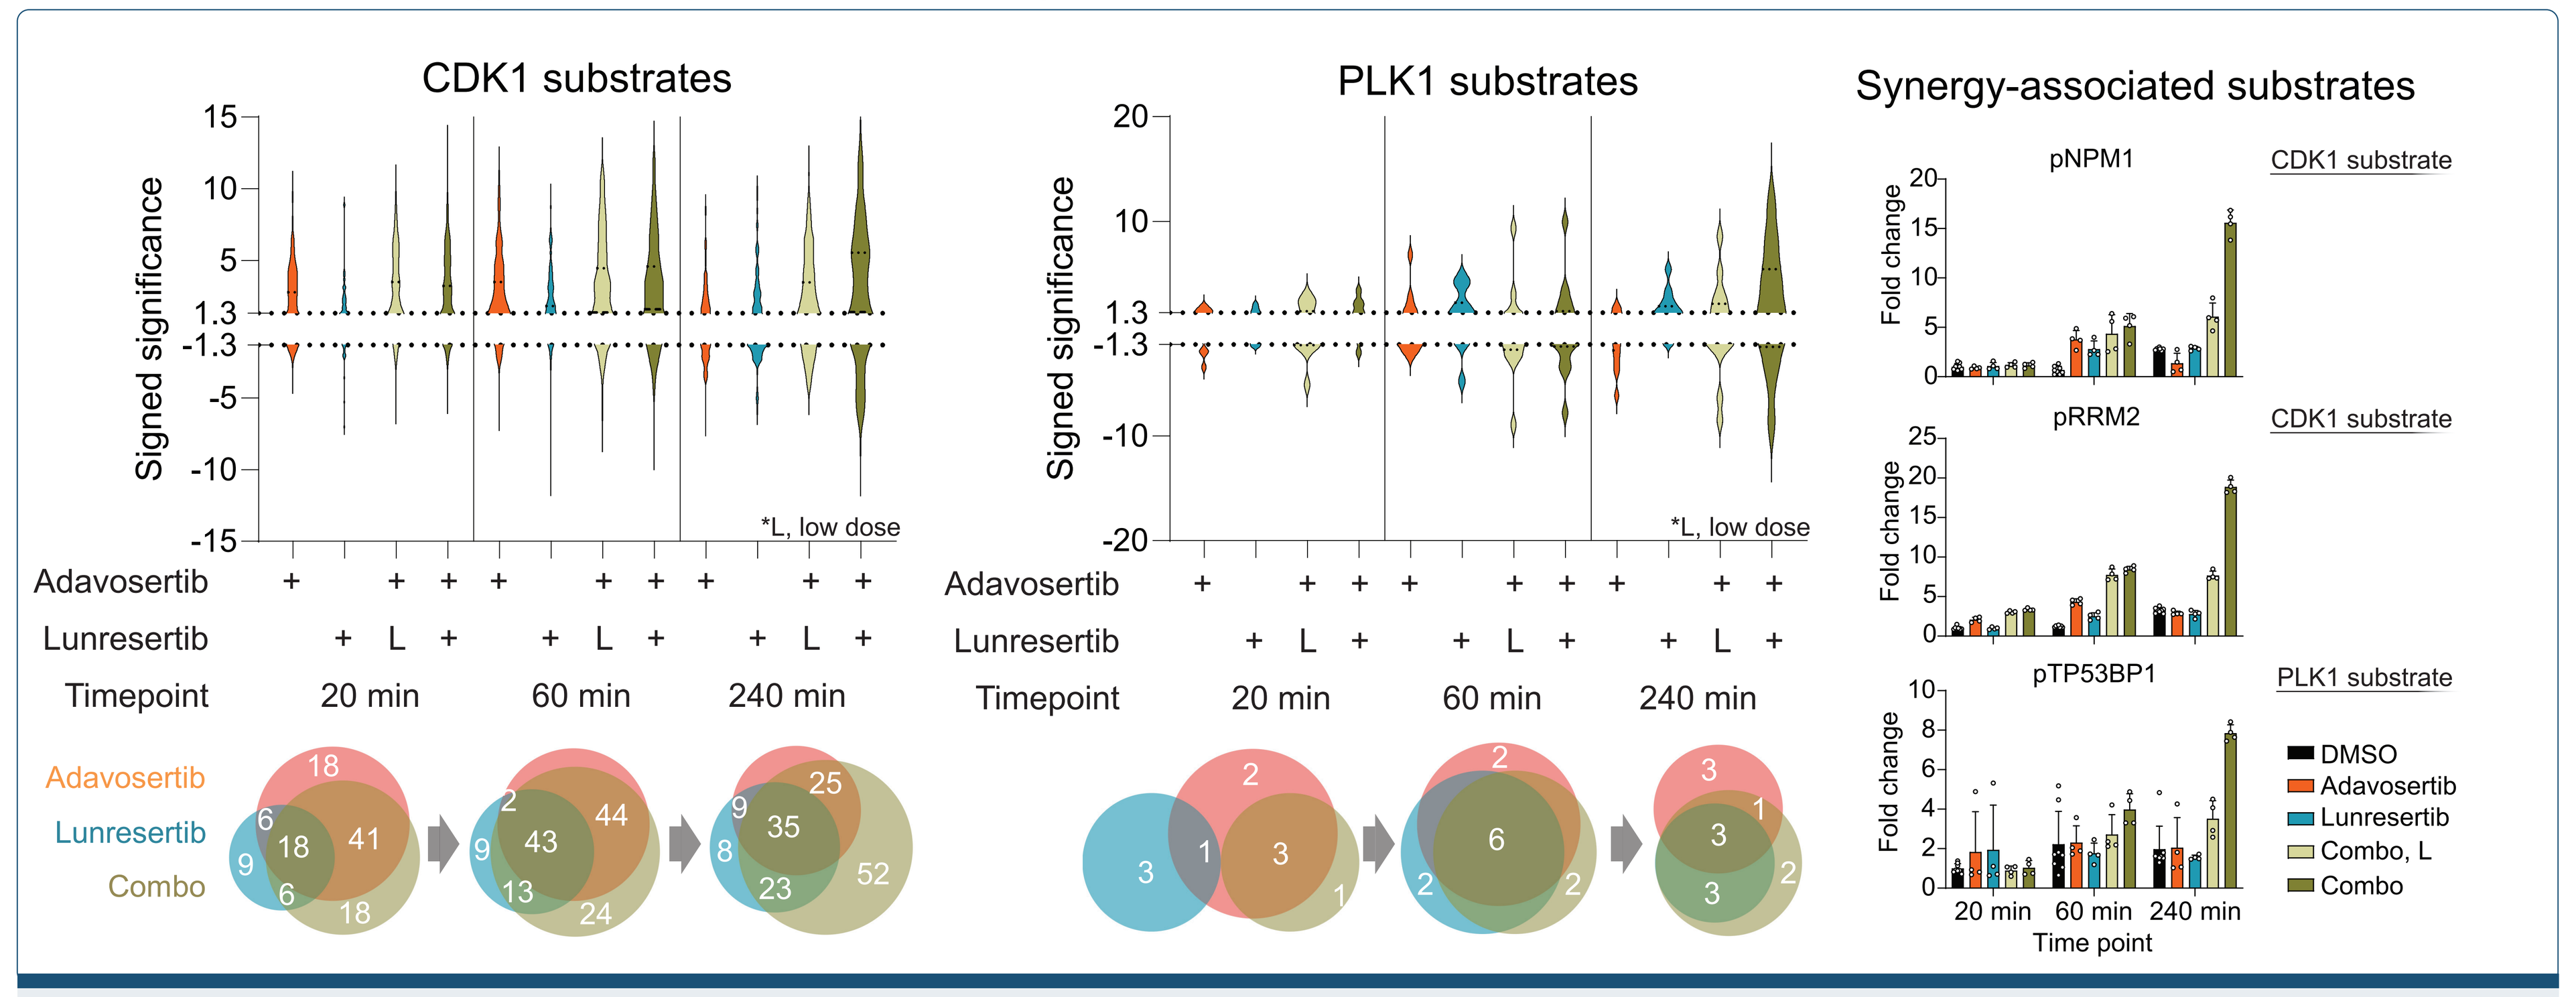
CDK1 substrates
PLK1 substrates
Synergy-associated substrates
Signed significance
*L, low dose
Adavosertib
Lunresertib
Timepoint
20 min
60 min
240 min
Combo
pNPM1
pRRM2
pTP53BP1
CDK1 substrate
PLK1 substrate
Fold change
Time point
DMSO
Combo, L

Mitotic biology: AP3 identifies mitotic phospho-substrates that define the synergistic biology of combined WEE1 and PKMYT1 inhibition.

AP3 analysis of desirable mitotic signaling altered by internal compounds

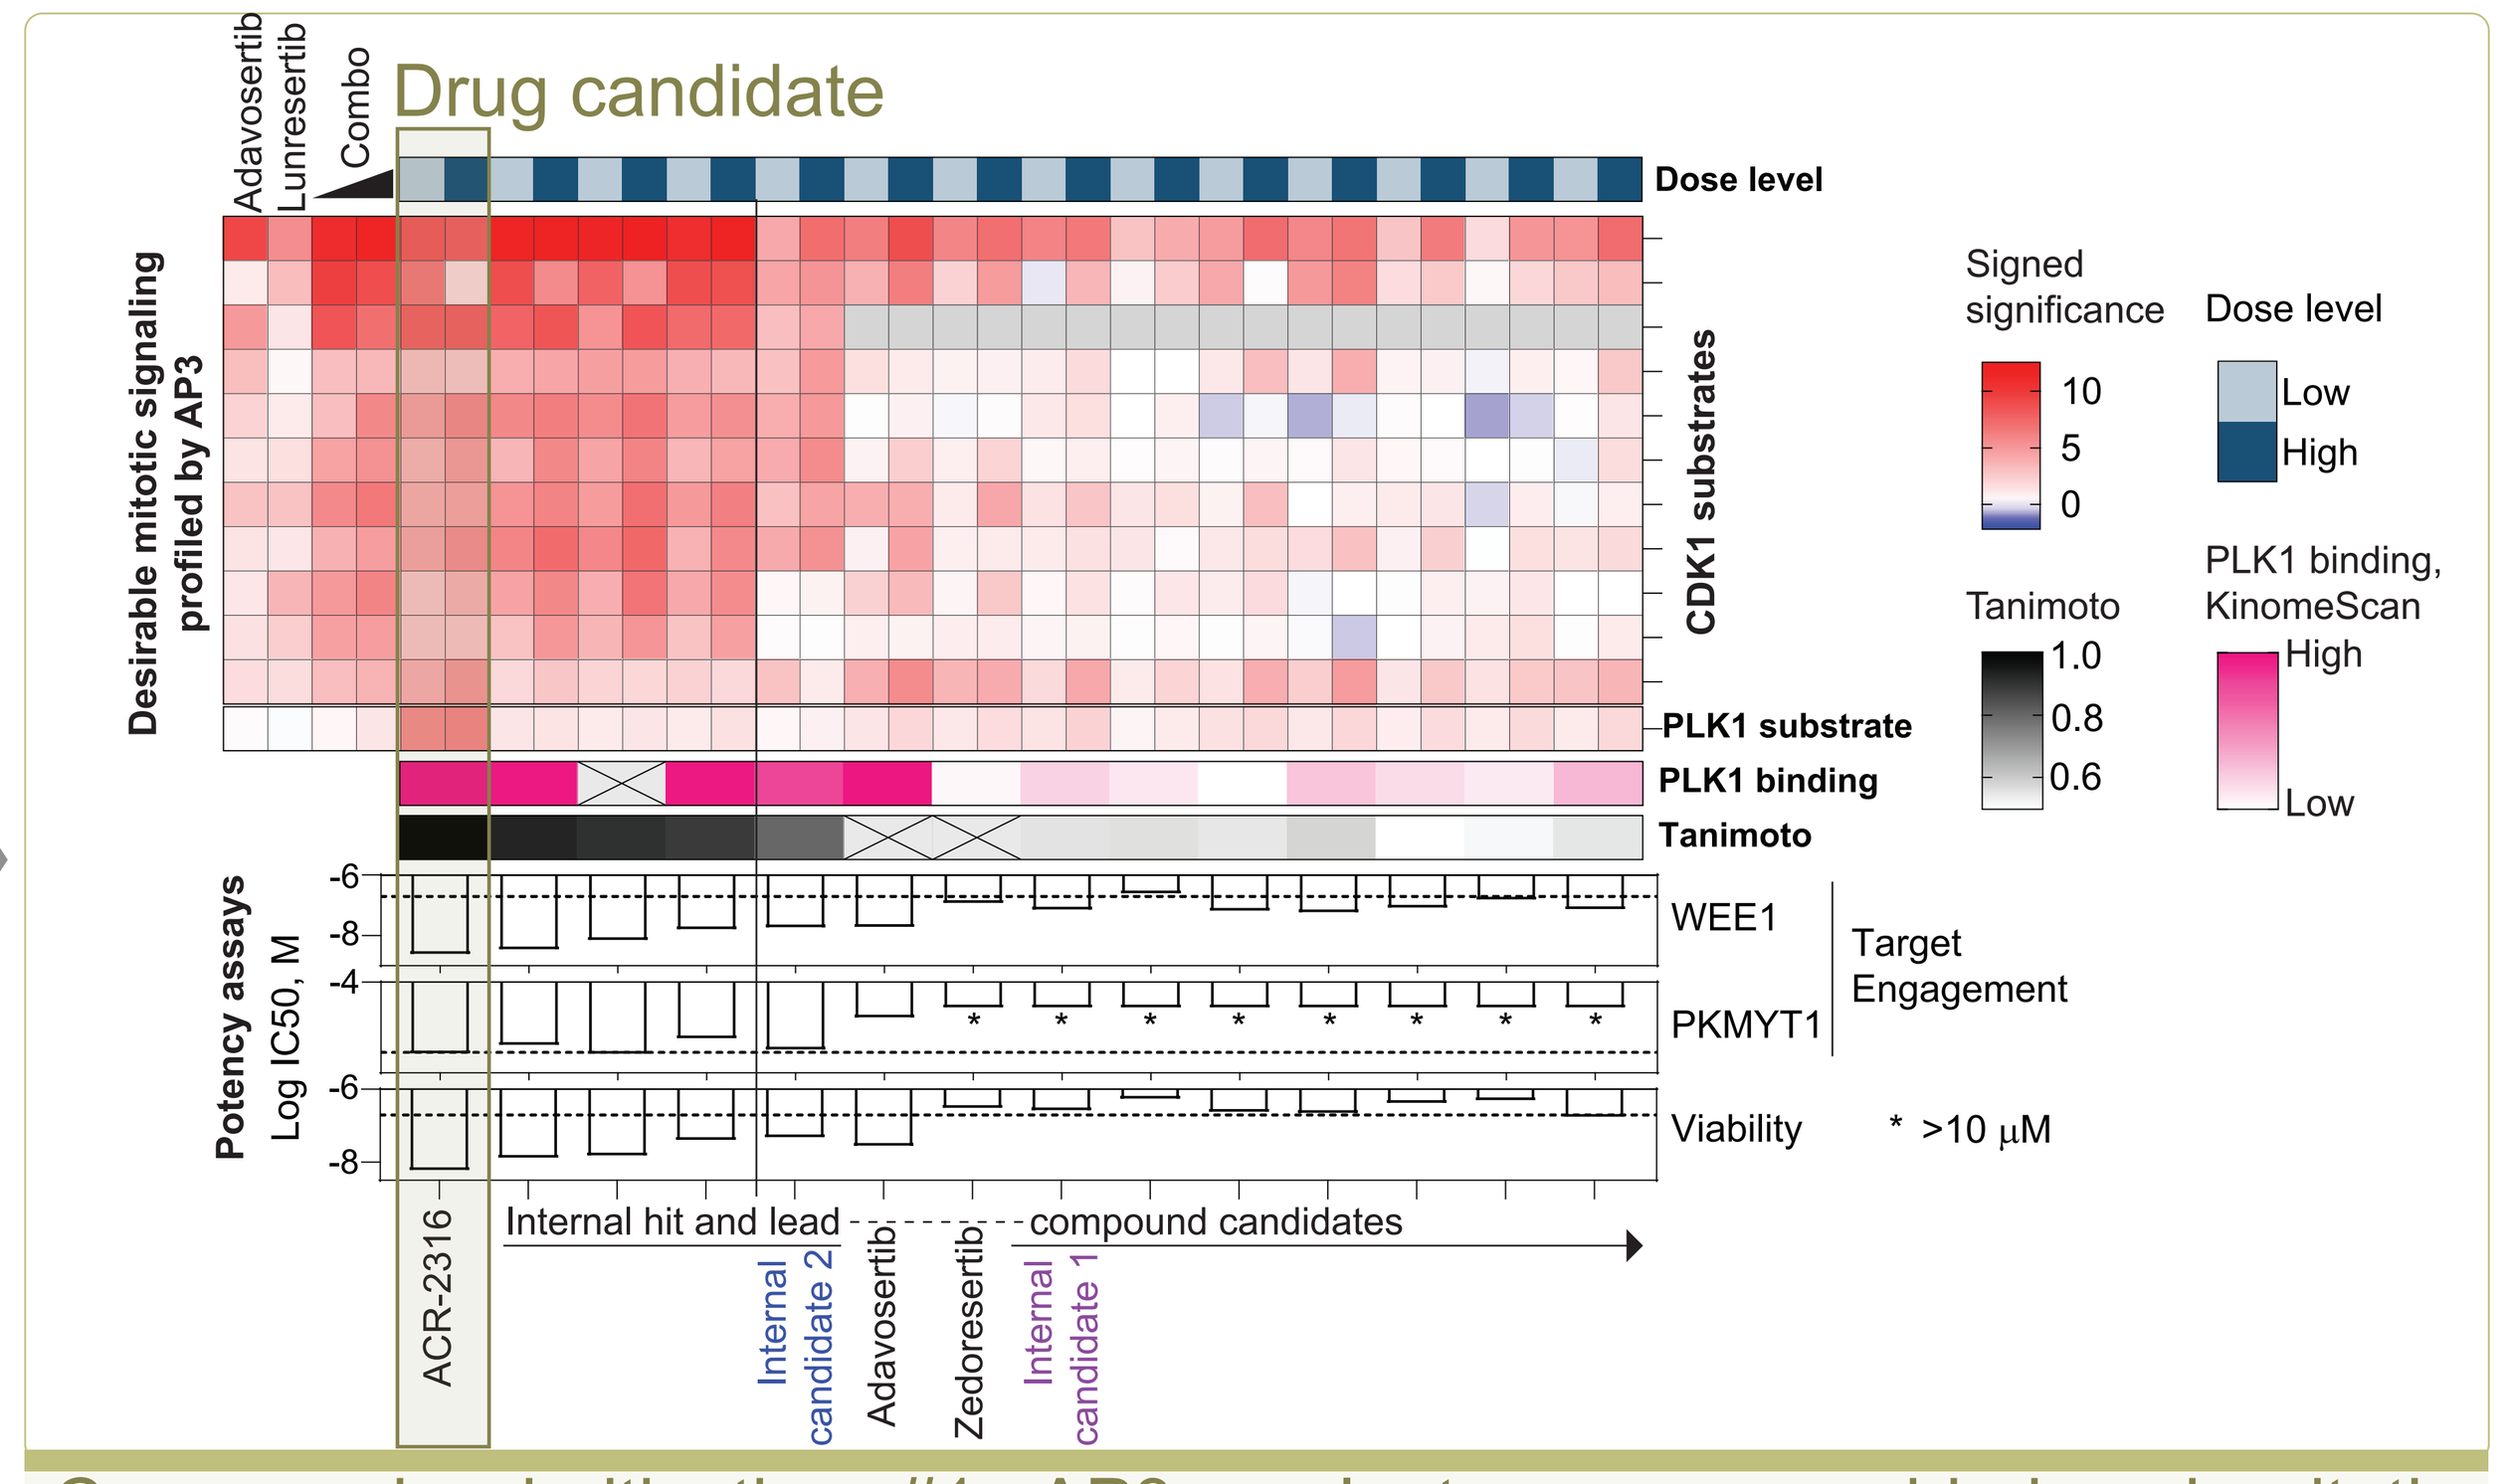
Drug candidate
Dose level
Desirable mitotic signaling profiled by AP3
CDK1 substrates
PLK1 substrate
PLK1 binding
Tanimoto
Potency assays Log IC50, M
WEE1
PKMYT1
Viability
Target Engagement
* >10 μM
Signed significance
Low
High
PLK1 binding, KinomeScan
Internal hit and lead compound candidates
ACR-2316
Internal candidate 2
Adavosertib
Zedoresertib
Internal candidate 1

Compound prioritization #1: AP3 evaluates compound-induced mitotic phosphorylation and prioritizes candidates that produce the desired signaling state, including potent PLK1 activation to ensure mitotic execution and pro-apoptotic cell death.

2. AP3 Biological SAR, Part 2: Mapping WEE1/PKMYT1 Phosphoproteomic Pathway Signatures Associated with Therapeutic Index to Guide Compound Selection

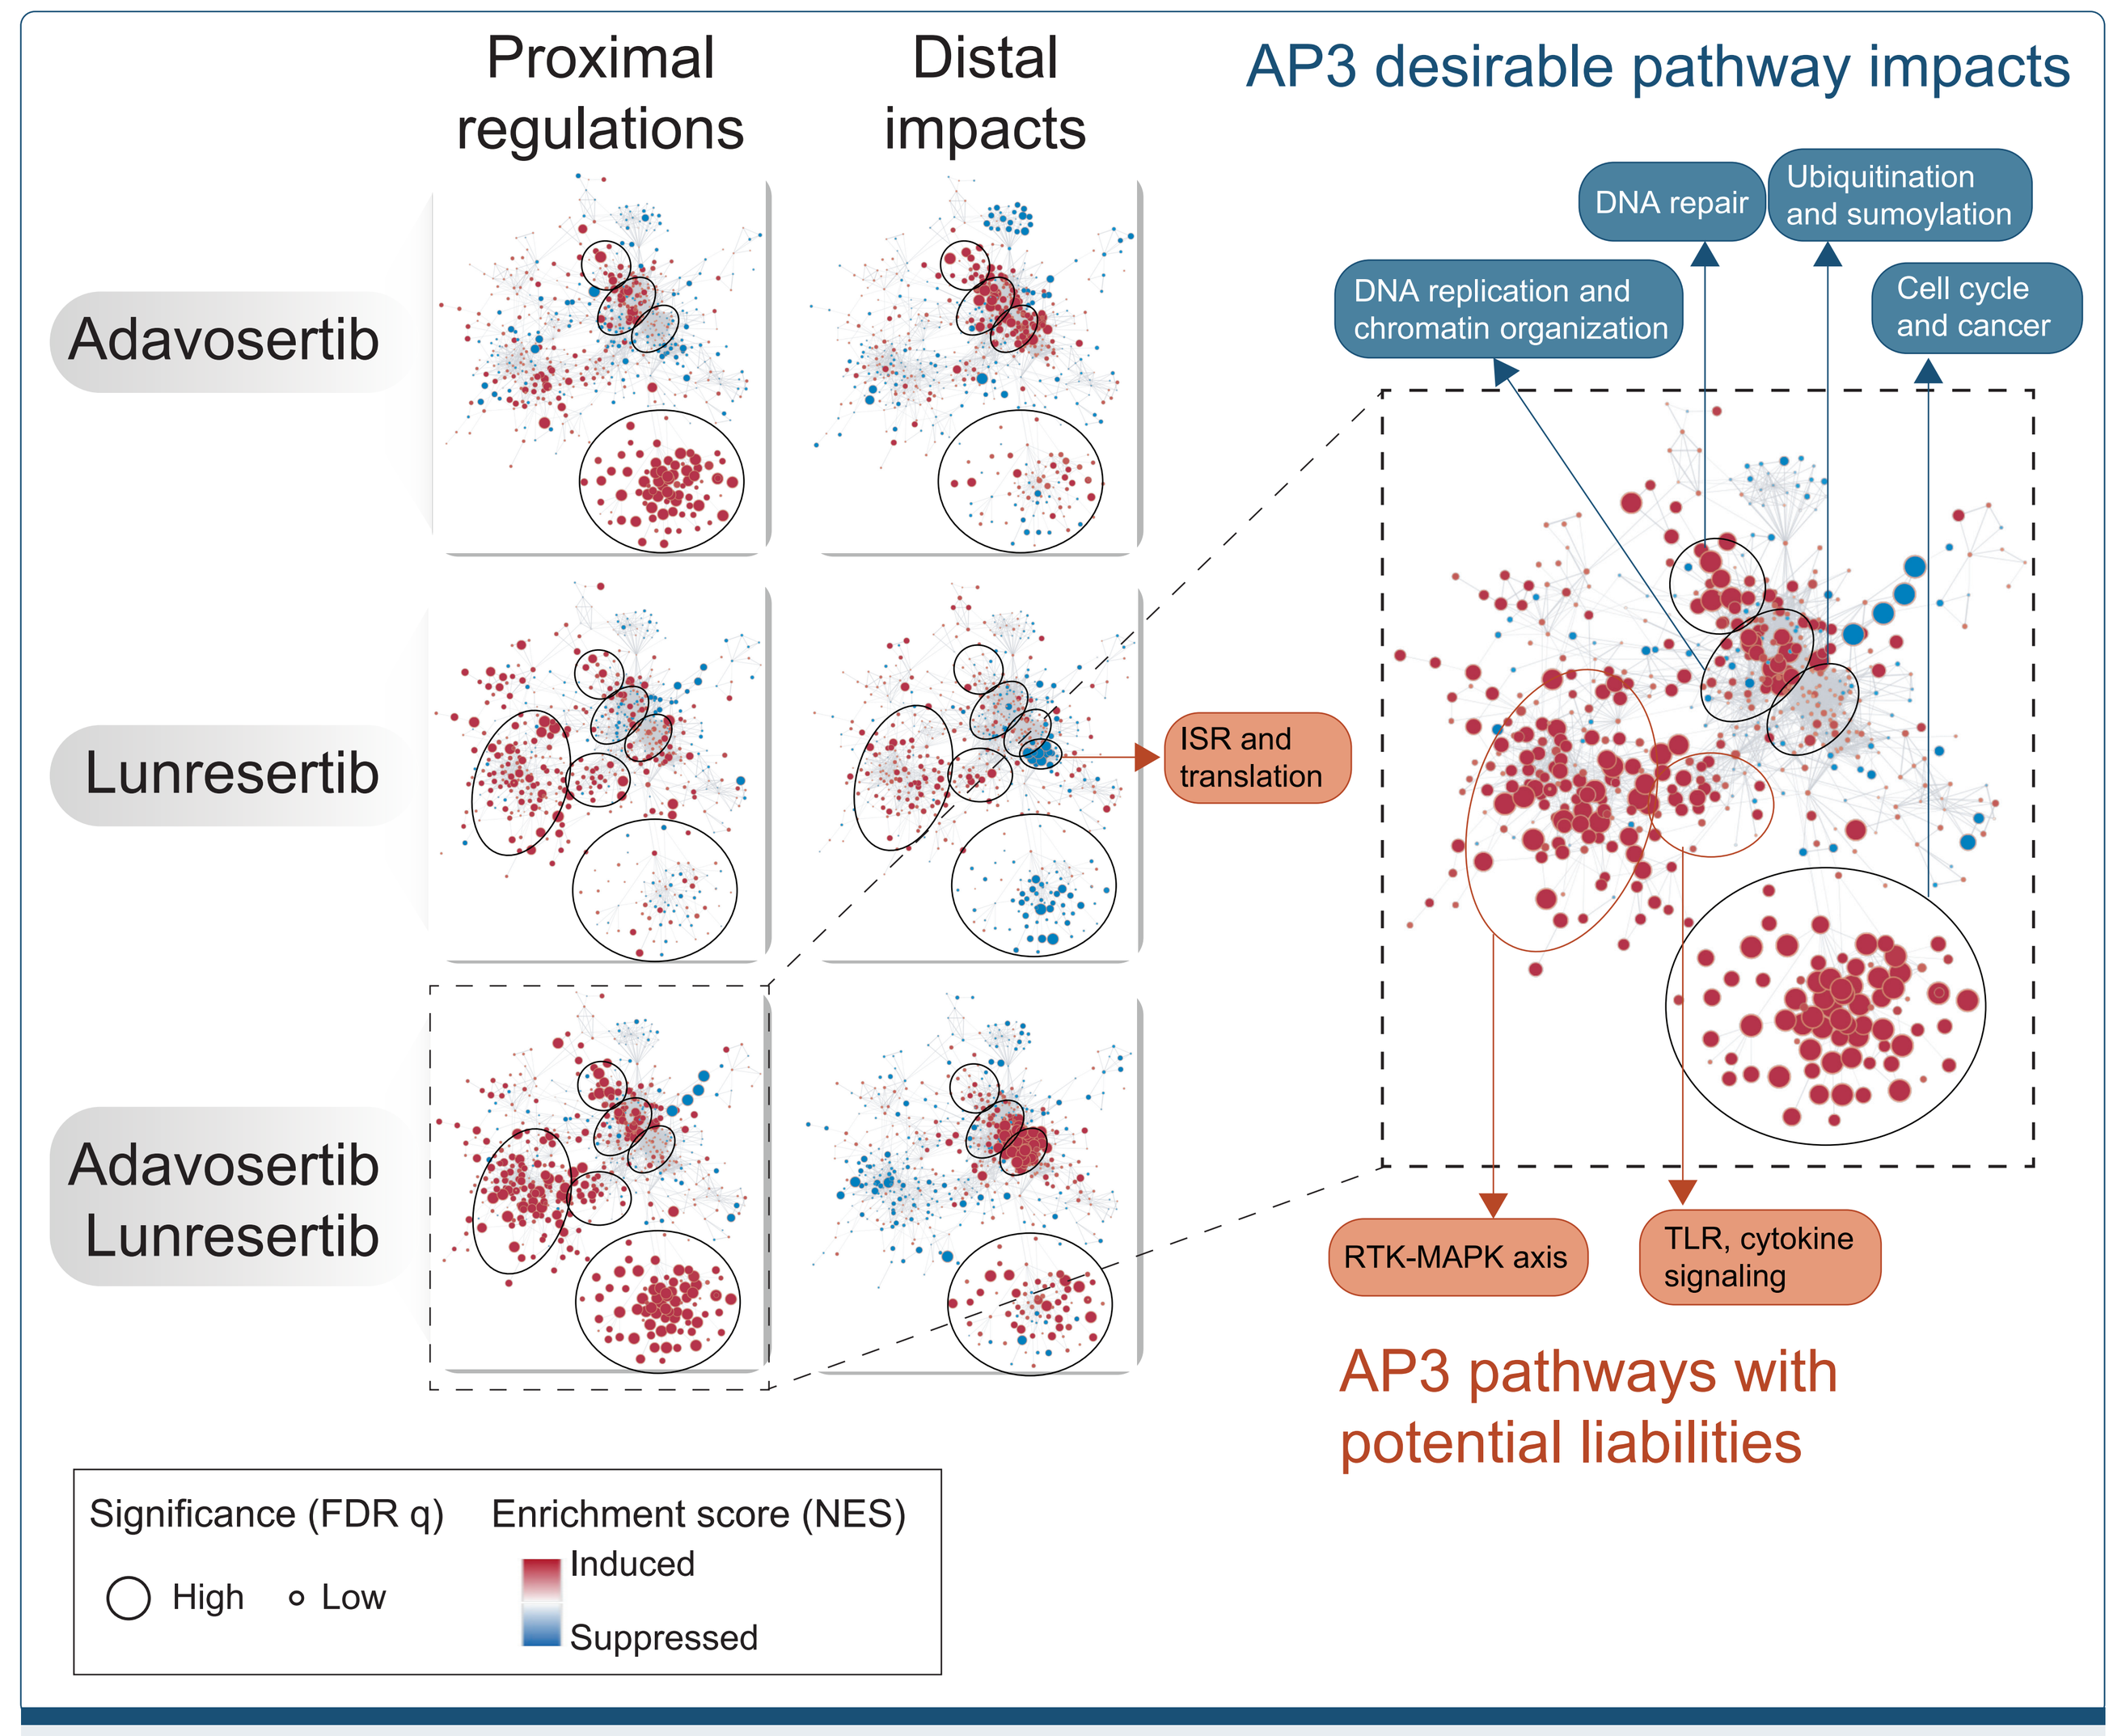
Proximal regulations
Distal impacts
AP3 desirable pathway impacts
Adavosertib
Lunresertib
Adavosertib Lunresertib
DNA repair
Ubiquitination and sumoylation
DNA replication and chromatin organization
Cell cycle and cancer
ISR and translation
RTK-MAPK axis
TLR, cytokine signaling
AP3 pathways with potential liabilities
Significance (FDR q)
High
Low
Enrichment score (NES)
Induced
Suppressed

Pathway mapping: AP3 defines the phosphoproteomic signaling regulated by single agent and combined WEE1/PKMYT1 inhibition, distinguishing desirable pathway effects from those associated with potential liabilities.

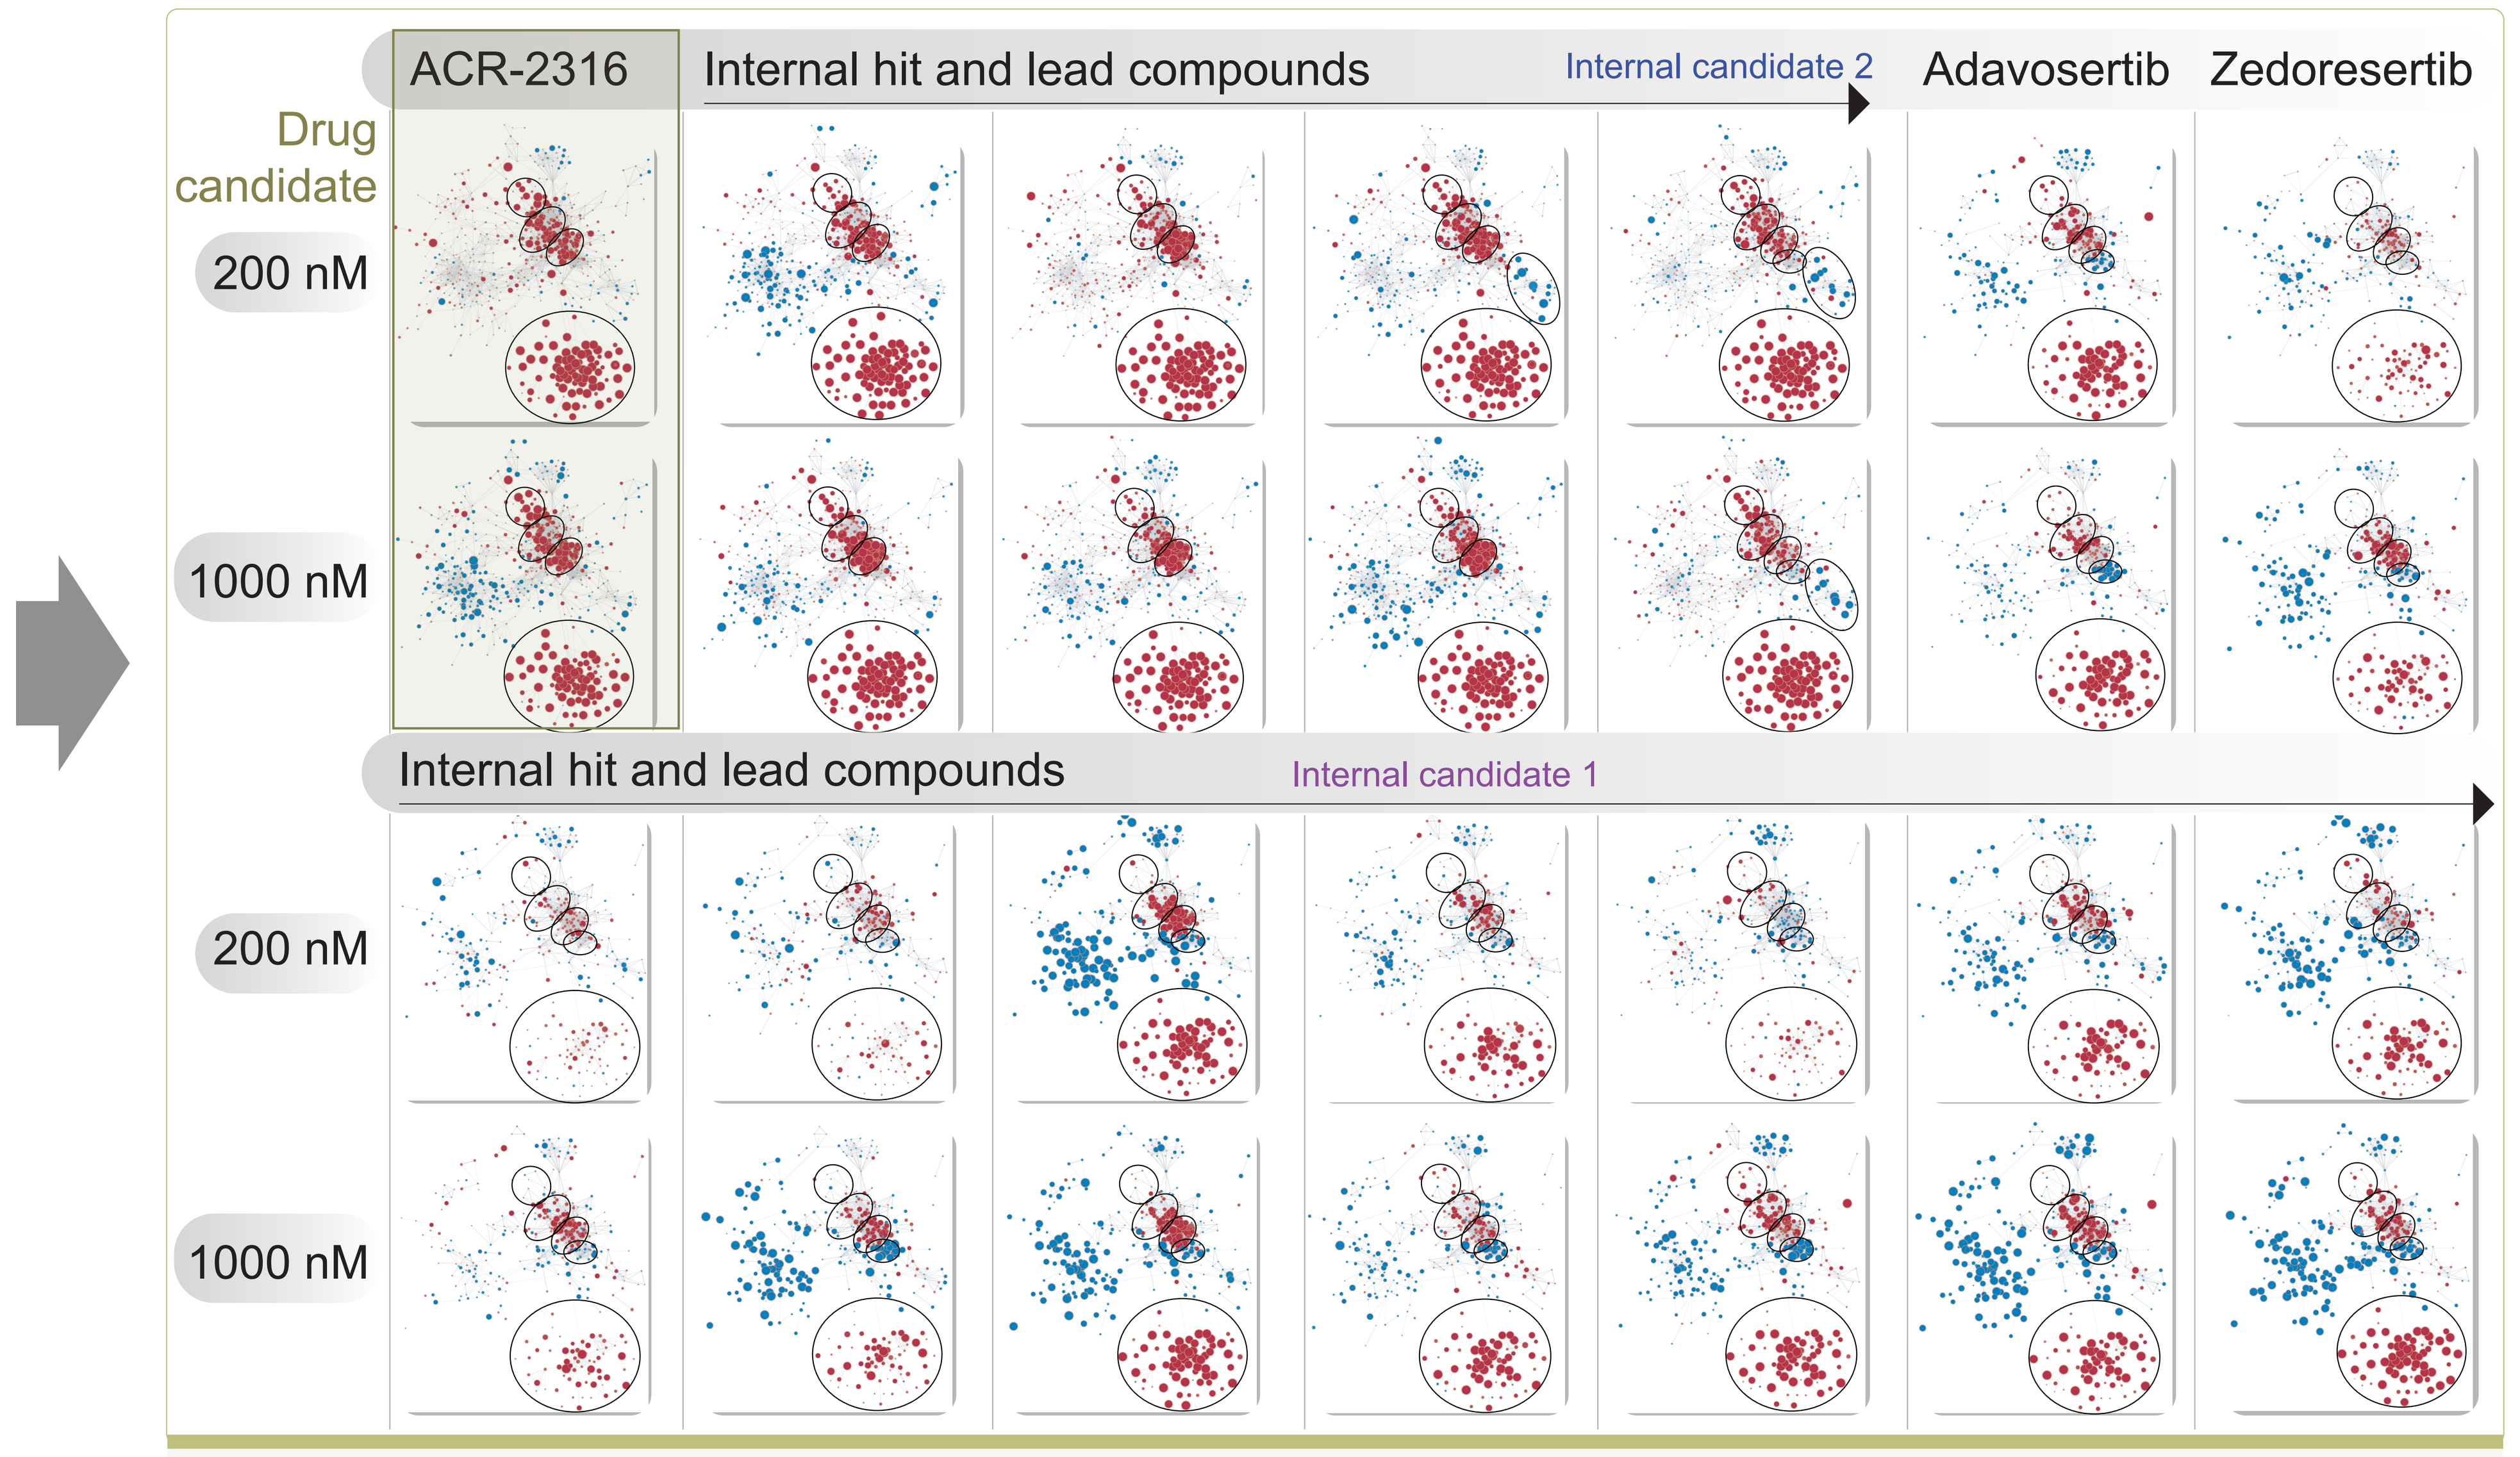
ACR-2316
Internal hit and lead compounds
Internal candidate 2
Adavosertib
Zedoresertib
Drug candidate
200 nM
1000 nM
Internal hit and lead compounds
Internal candidate 1
200 nM
1000 nM

Compound prioritization #2: AP3 profiles phosphoproteomic signaling across internal development candidates, enabling compound prioritization based on established desirable pathway signatures.

RESULTS

3. AP3 characterization of DC candidate - ACR-2316

AP3 validates desirable mitotic signaling events induced by ACR-2316

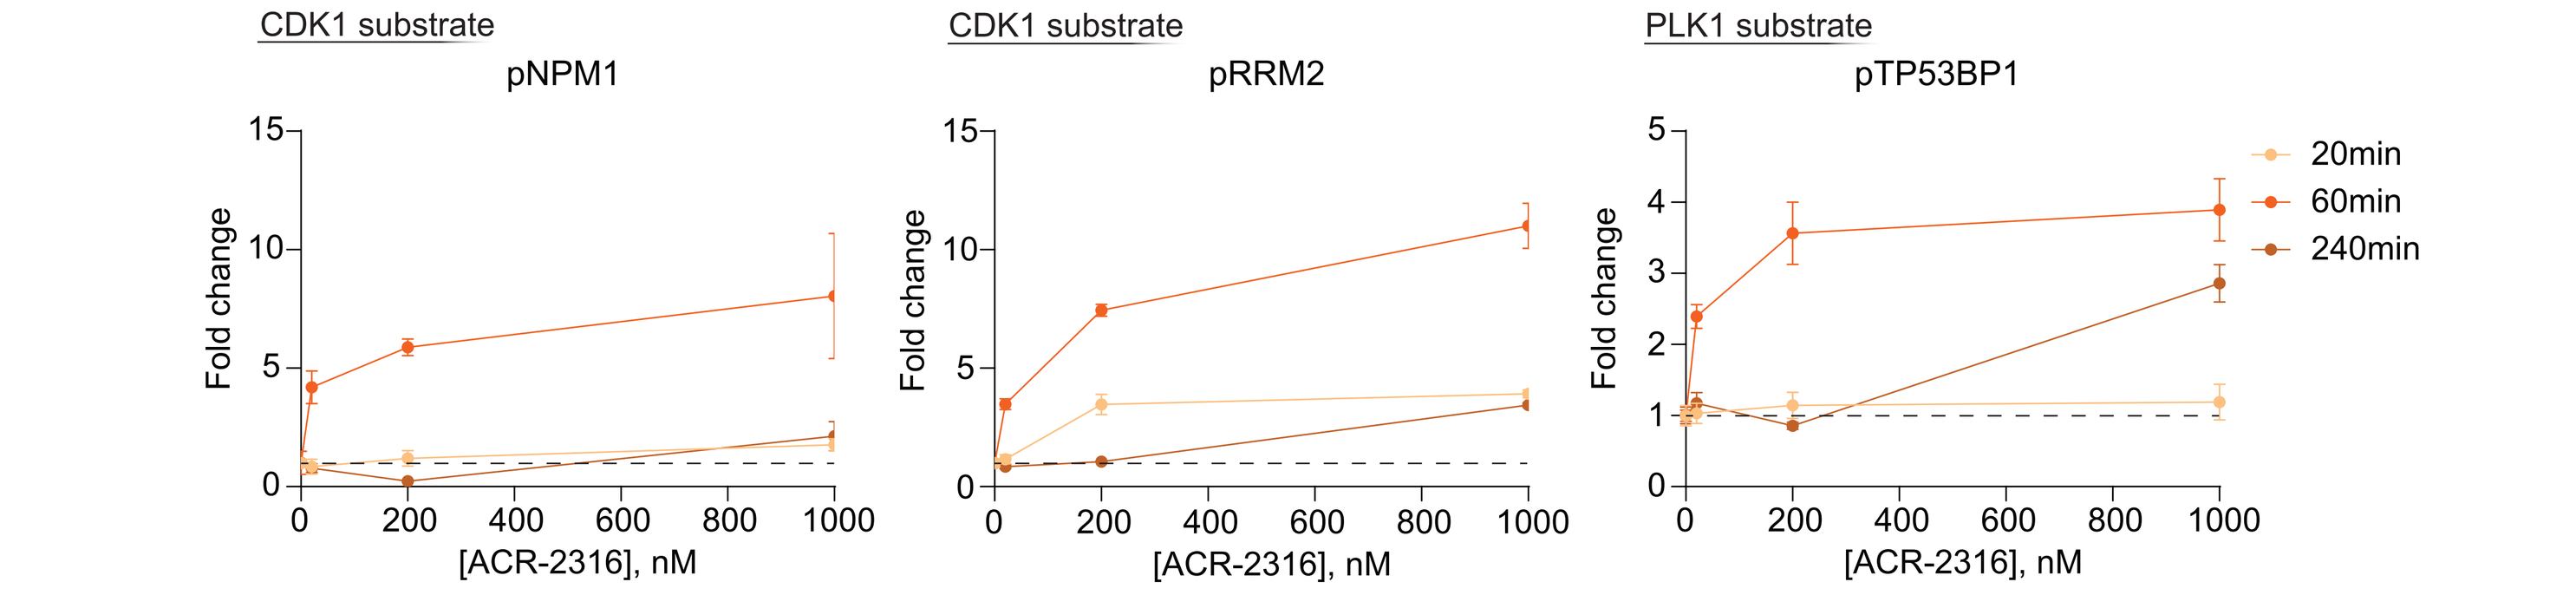
CDK1 substrate
pNPM1
CDK1 substrate
pRRM2
PLK1 substrate
pTP53BP1
Fold change
[ACR-2316], nM
20min
60min
240min

AP3 validates desirable pathway signaling effects regulated by ACR-2316

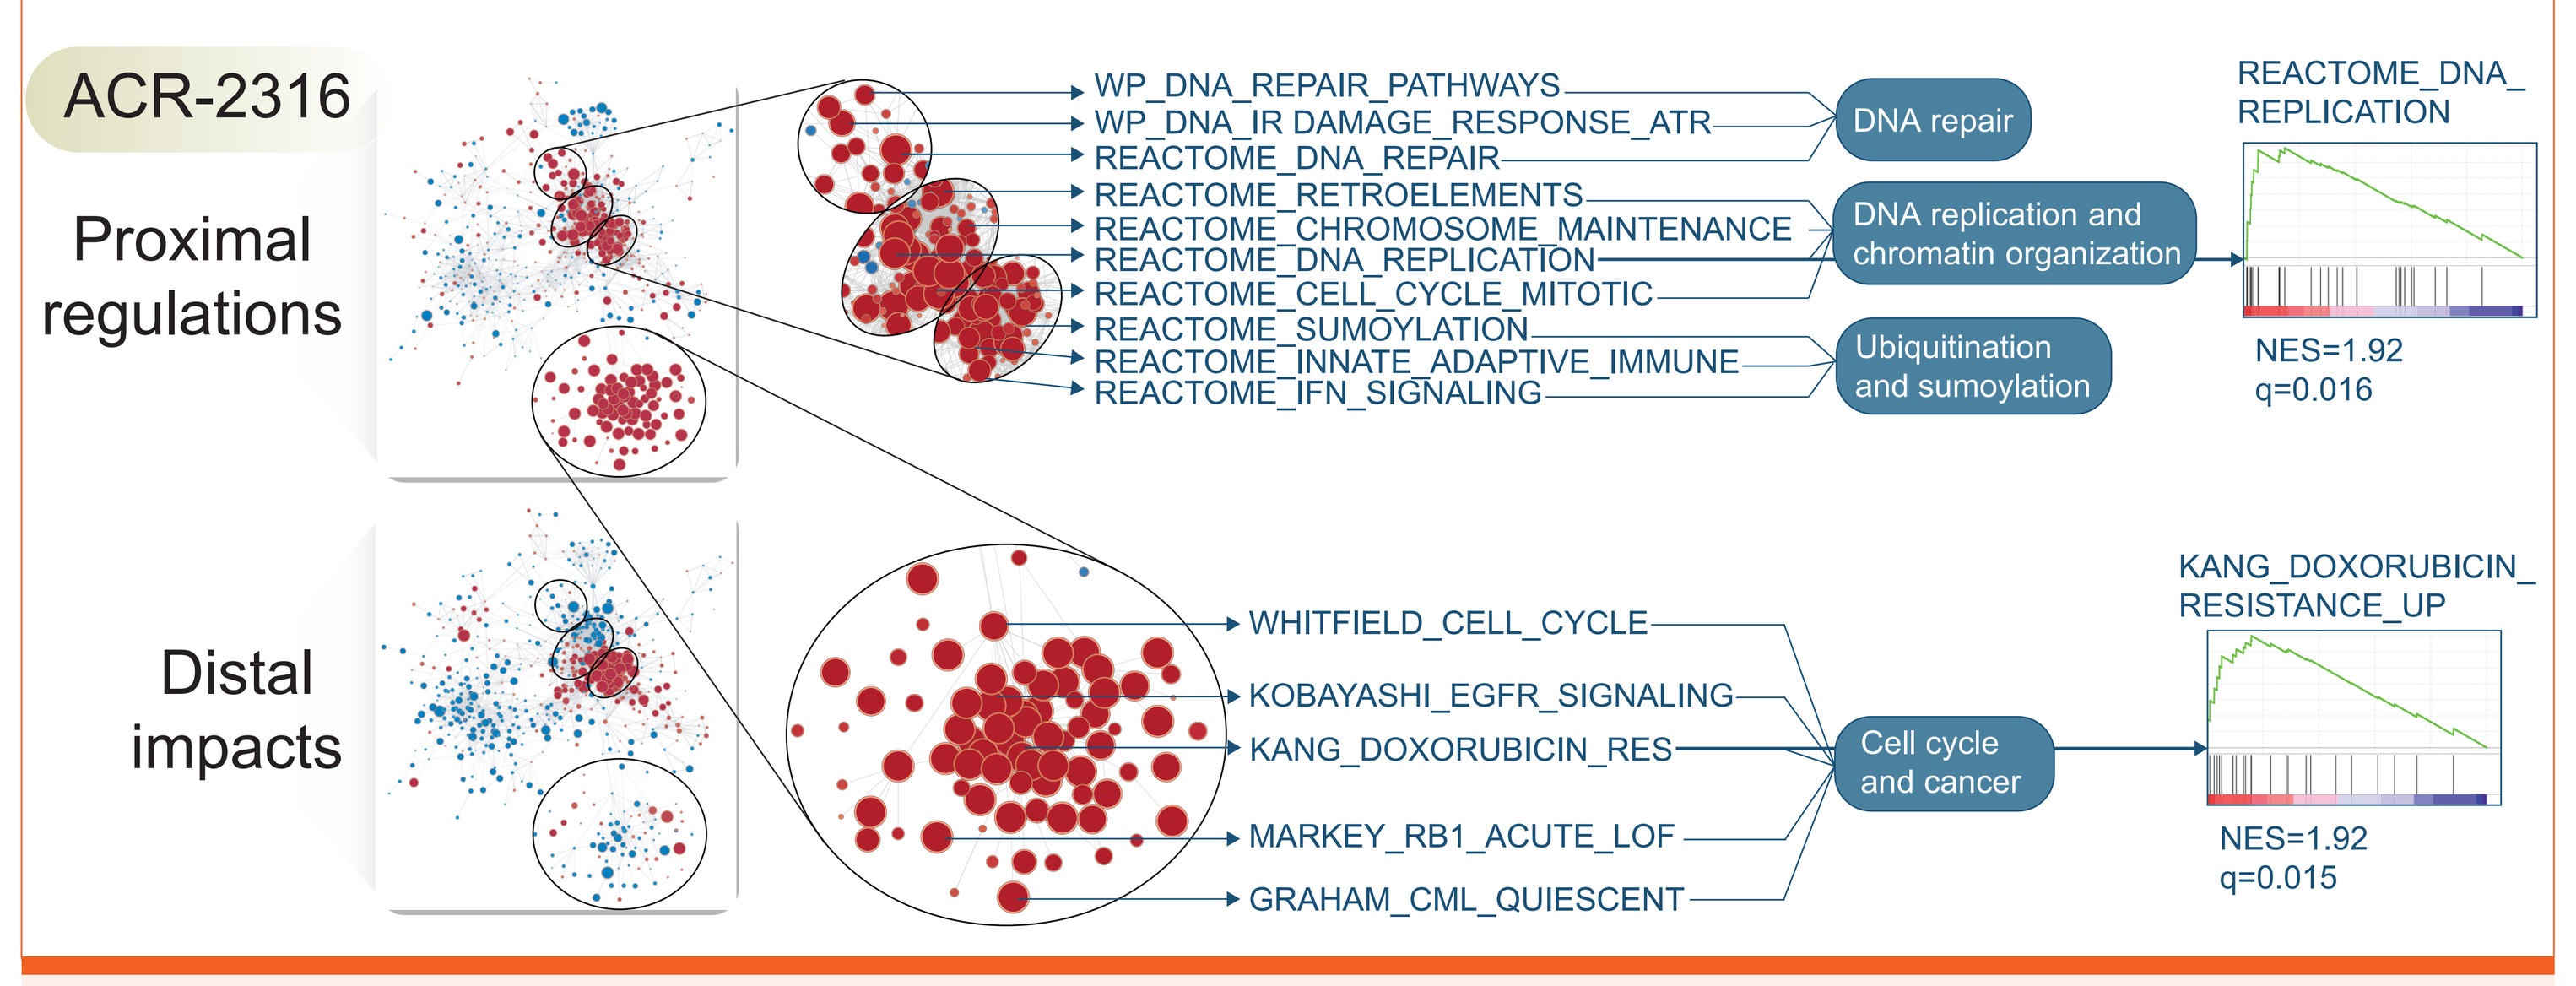
ACR-2316
Proximal regulations
Distal impacts
WP_DNA_REPAIR_PATHWAYS
WP_DNA_IR DAMAGE_RESPONSE_ATR
REACTOME_DNA_REPAIR
REACTOME_RETROELEMENTS
REACTOME_CHROMOSOME_MAINTENANCE
REACTOME_DNA_REPLICATION
REACTOME_CELL_CYCLE_MITOTIC
REACTOME_SUMOYLATION
REACTOME_INNATE_ADAPTIVE_IMMUNE
REACTOME_IFN_SIGNALING
DNA repair
DNA replication and chromatin organization
Ubiquitination and sumoylation
REACTOME_DNA_REPLICATION
NES=1.92
q=0.016
WHITFIELD_CELL_CYCLE
KOBAYASHI_EGFR_SIGNALING
KANG_DOXORUBICIN_RES
MARKEY_RB1_ACUTE_LOF
GRAHAM_CML_QUIESCENT
Cell cycle and cancer
KANG_DOXORUBICIN_RESISTANCE_UP
NES=1.92
q=0.015

DC characterization: AP3 comprehensively profiles ACR-2316 across dose levels and time points, confirming robust regulation of CDK1, PLK1, and the desired phosphoproteomic pathway signatures.

4. *In vivo* efficacy: ACR-2316 demonstrates the greatest antitumor activity among internal candidates in a human lung tumor xenograft model and induces robust CDK1 and PLK1 signaling.

ACR-2316 demonstrates superior antitumor efficacy compared with internal candidates lacking the desired AP3-defined pathway

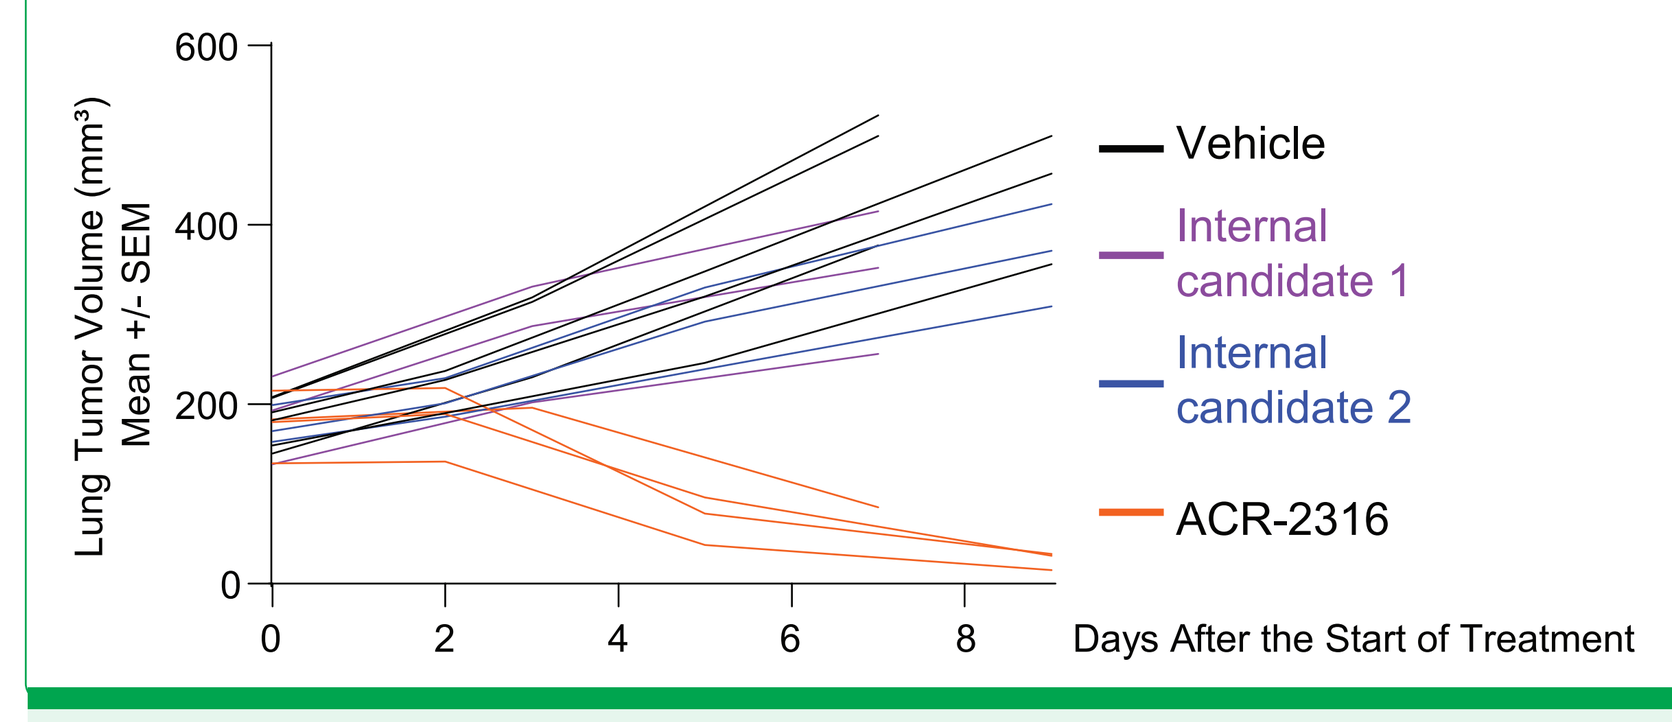
Lung Tumor Volume (mm³) Mean +/- SEM
Days After the Start of Treatment
Vehicle
Internal candidate 1
Internal candidate 2
ACR-2316

mIF validates that ACR-2316 most effectively induces the desired mitotic events identified by AP3

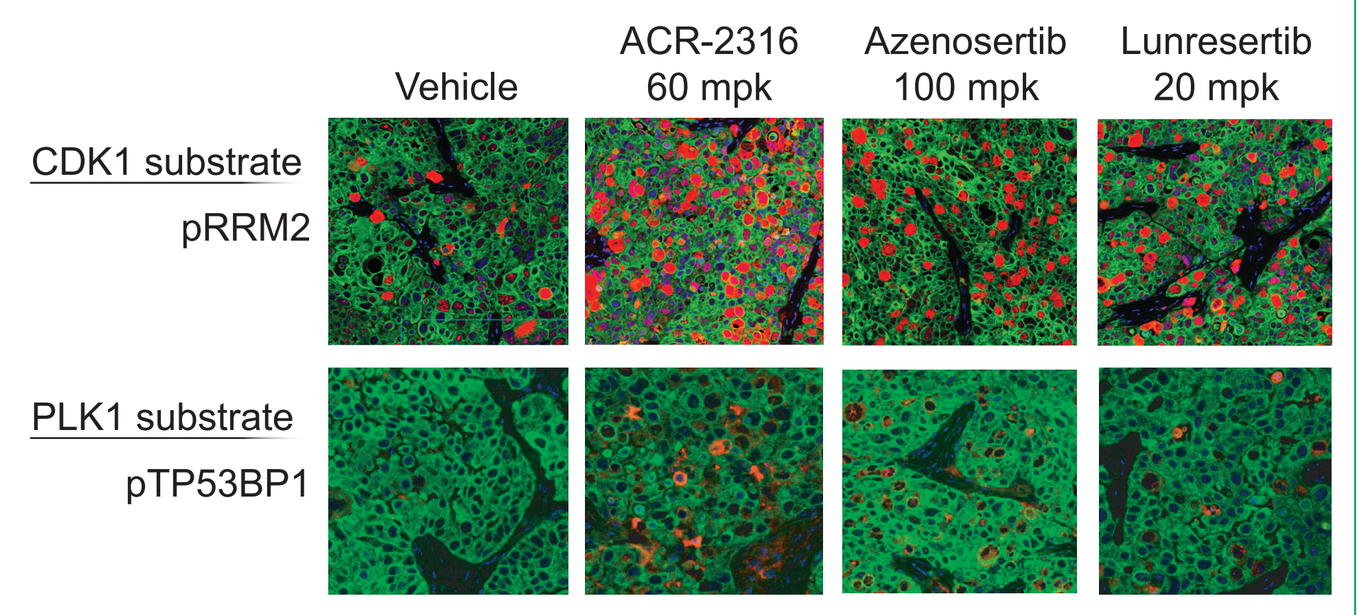
Vehicle
ACR-2316 60 mpk
Azenosertib 100 mpk
Lunresertib 20 mpk
CDK1 substrate
pRRM2
PLK1 substrate
pTP53BP1

AP3 SAR validation: Experimental validation confirmed AP3-guided nomination of ACR-2316 and established mitotic pharmacodynamic biomarkers for continued clinical development.

REFERENCES

- AACR Annual Meeting, 2024. *Cancer Res (2024) 84 (6_Supplement): 1977.*
- AACR Annual Meeting, 2025. *Cancer Res (2025) 85 (8_Supplement_1): 357.*
- AACR-NCI-EORTC International Conference, 2025. *Mol Cancer Ther (2025) 24 (10_Supplement): A097.*
- AACR-NCI-EORTC International Conference, 2025. *Mol Cancer Ther (2025) 24 (10_Supplement): C112.*
- AACR Annual Meeting, 2026. *Cancer Res (2026) 86 (7_Supplement): 3789.*

Prior ACR-2316 AACR publications

ACRIVON THERAPEUTICS